Statistical characterization of medical images of bone

Elena Ajayi [1,2] and Jonathan D. Victor [2]

[1] St. John's University, [2] Weill Cornell Medical College NY, NY

VSS2023
53.414

Introduction

- Visual perception has evolved to process natural images.
- Medical images, however, are formed by different physical processes than natural images and therefore may have different characteristics.
- We investigated the extent of these differences in two classes of medical images: bone radiographs and scintigrams.
- For each, we determined the power spectrum and local image statistics.

Methods

We analyzed two kinds of bone images: radiographs and scintigrams, selected from MedPix (https://medpix.nlm.nih.gov). Images included studies of extremities, cranium, spine, and thorax, and, for scintigrams, whole-body, and included studies of normal bone and a range of pathology.

Images were hand-curated to remove all artifacts and labeling, and then partitioned into rectangular regions of interest (ROI's). ROI's were then randomly subdivided into square patches of size 128 x 128 pixels, or smaller (see pipeline).

Radiographs

- 51 source images
- 67 ROI's (1 to 3 per image)
- ROI width: 336 to 1033 pixels
- ROI height: 374 to 2021 pixels
- 1006 patches (128 x 128 pixels), 362 entirely interior

Scintigrams

- 23 source images
- 73 ROI's (1 to 6 per image)
- ROI width: 230 to 1024 pixels
- ROI height: 134 to 1200 pixels
- 188 patches (128 x 128 pixels)

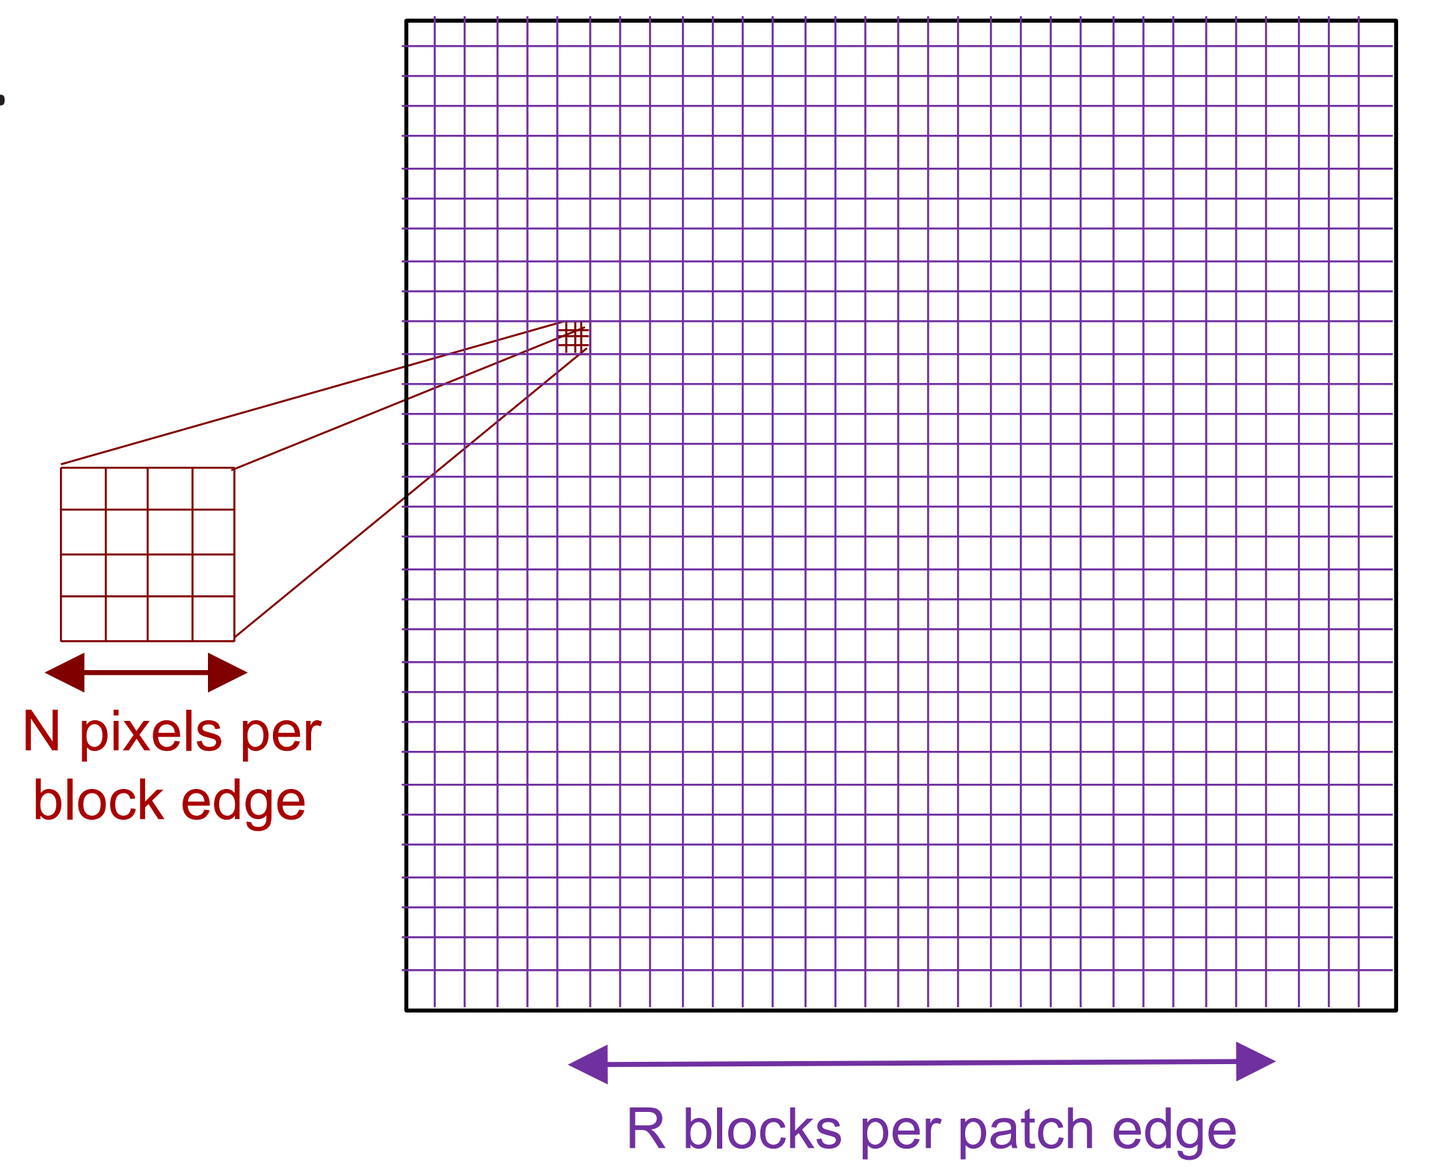

N pixels per block edge
R blocks per patch edge

After curation, images were subdivided into patches of size 128 x 128 pixels, downsampled in blocks of N (N=1, 2, 4, 8, or 16), and then analyzed in patches of R x R blocks (R=16, 32, 64, or 128), with NR <=128.

Image Formation

Natural images and medical images are formed differently. Therefore, we anticipate that their respective statistics will differ.

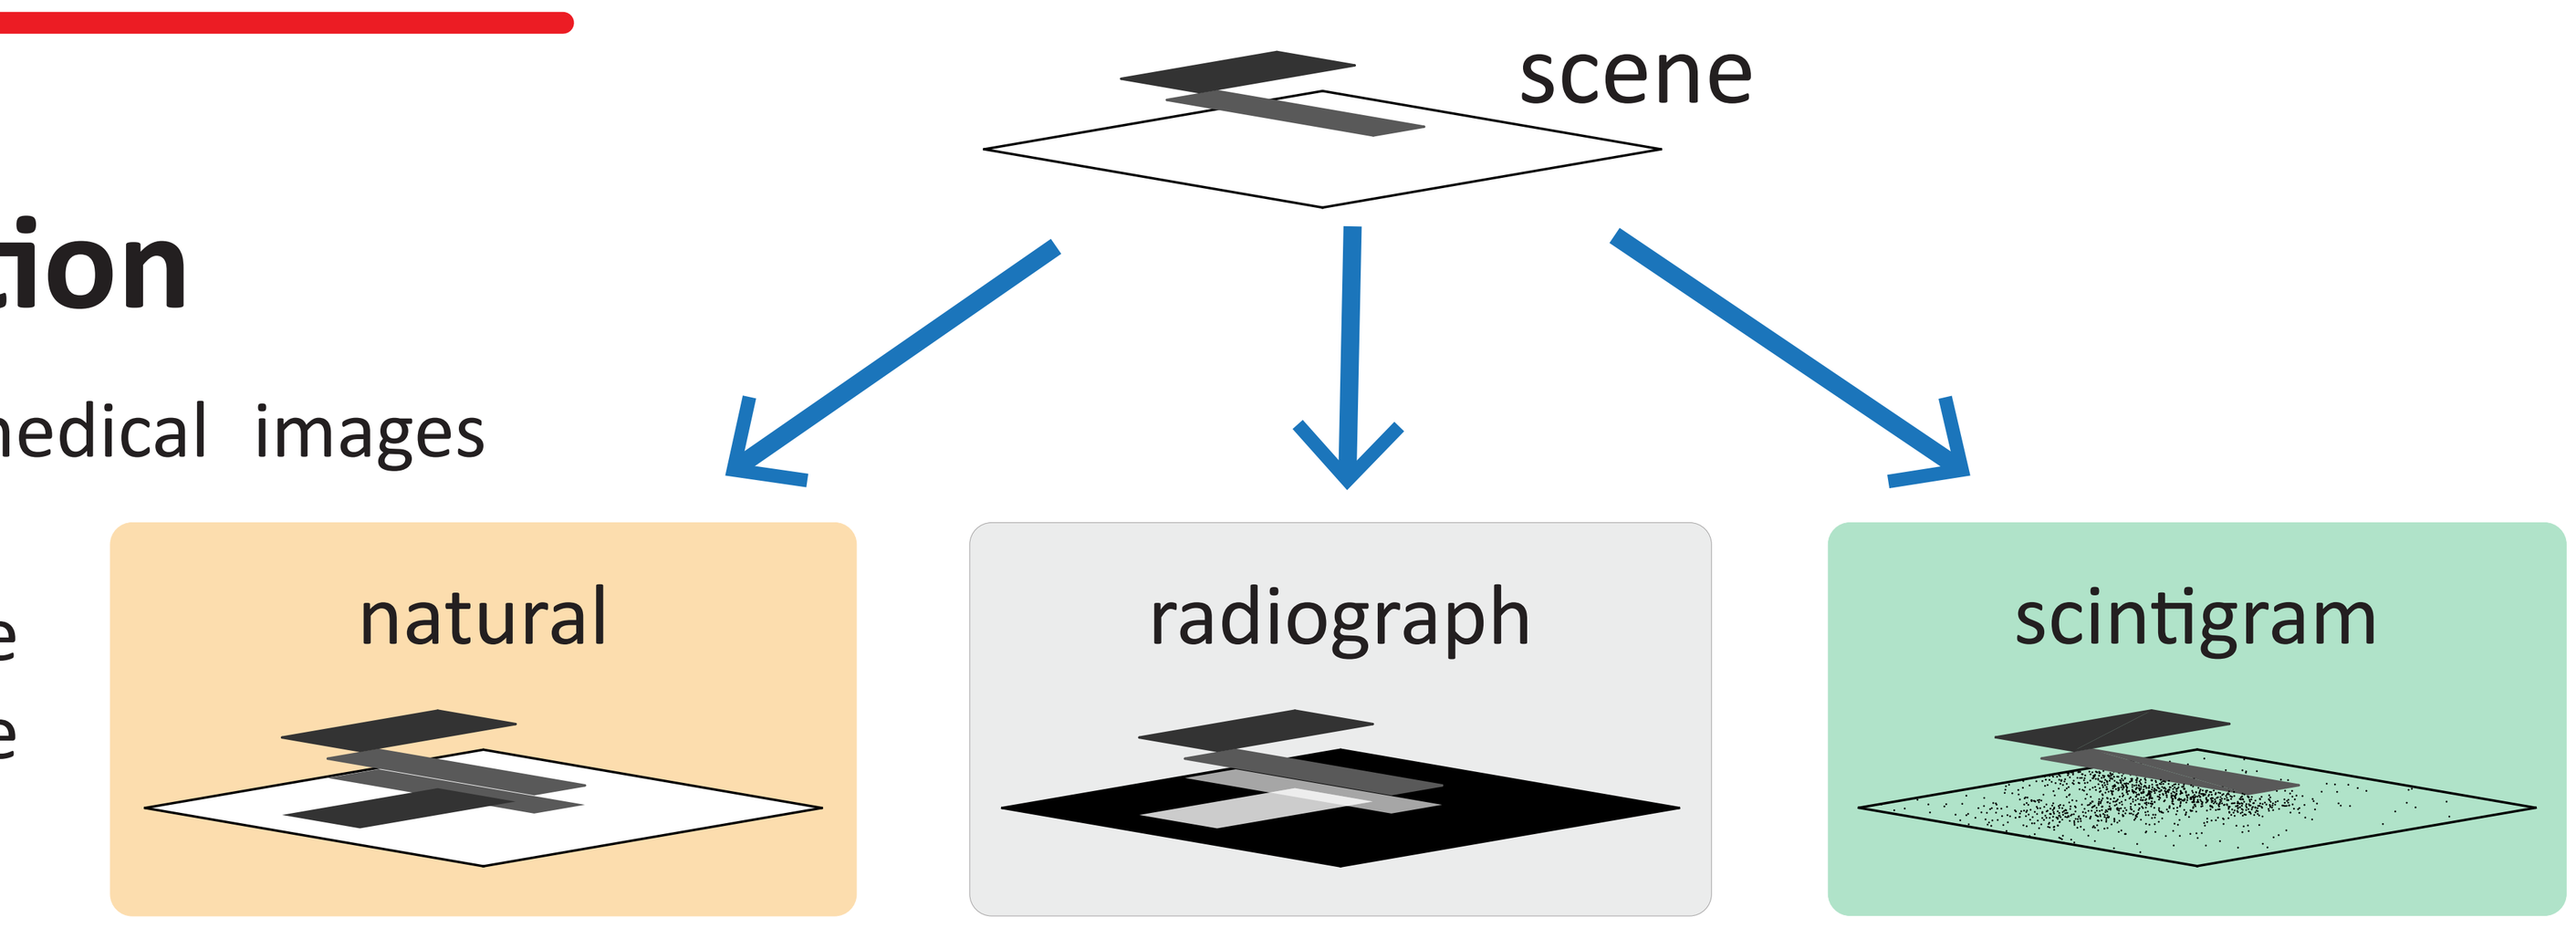

scene
natural
radiograph
scintigram

Natural images are typically formed by interaction of light with opaque objects. This will generate occlusions and T junctions.

Radiographs are formed by xrays passing through objects. This will generate transparency rather than occlusion.

In scintigrams, images are formed by decaying radioactive particles scattering in all directions from the object. Thus, there are no sharp edges.

Image Analysis Pipeline

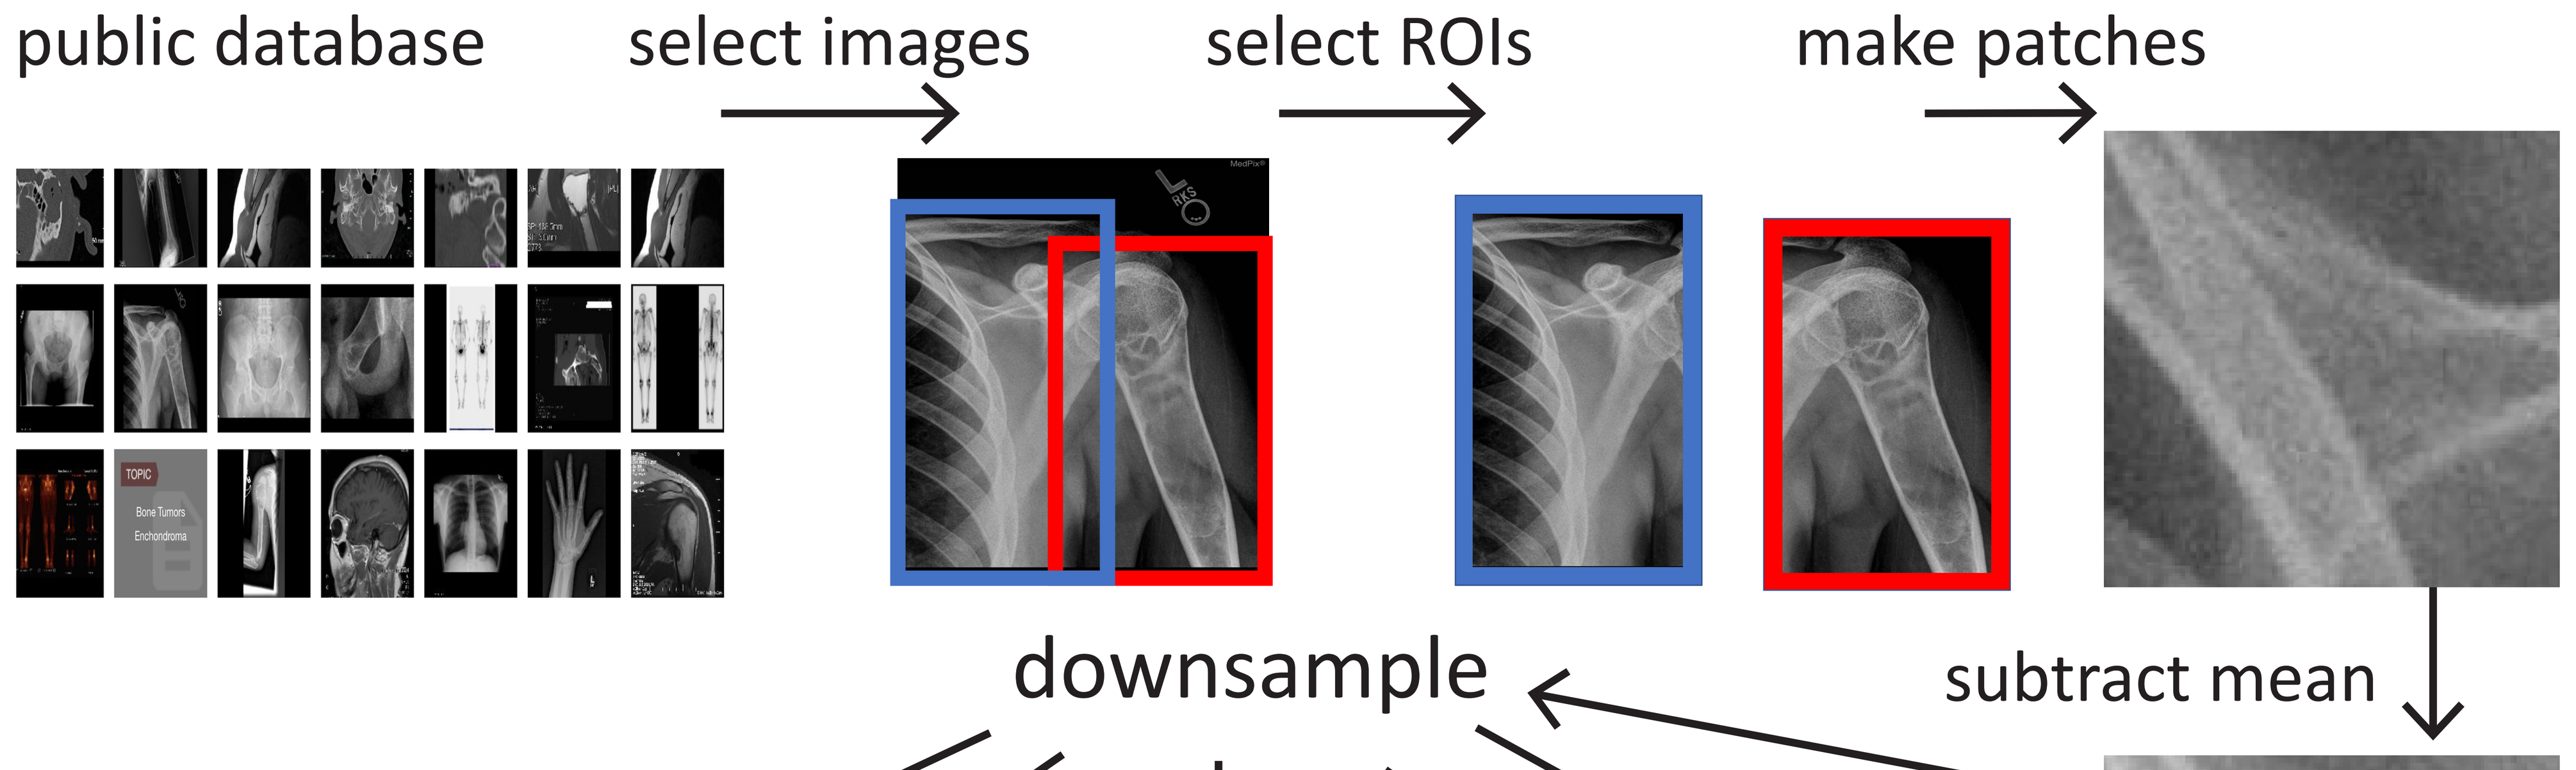

public database
select images
select ROIs
make patches
downsample
subtract mean

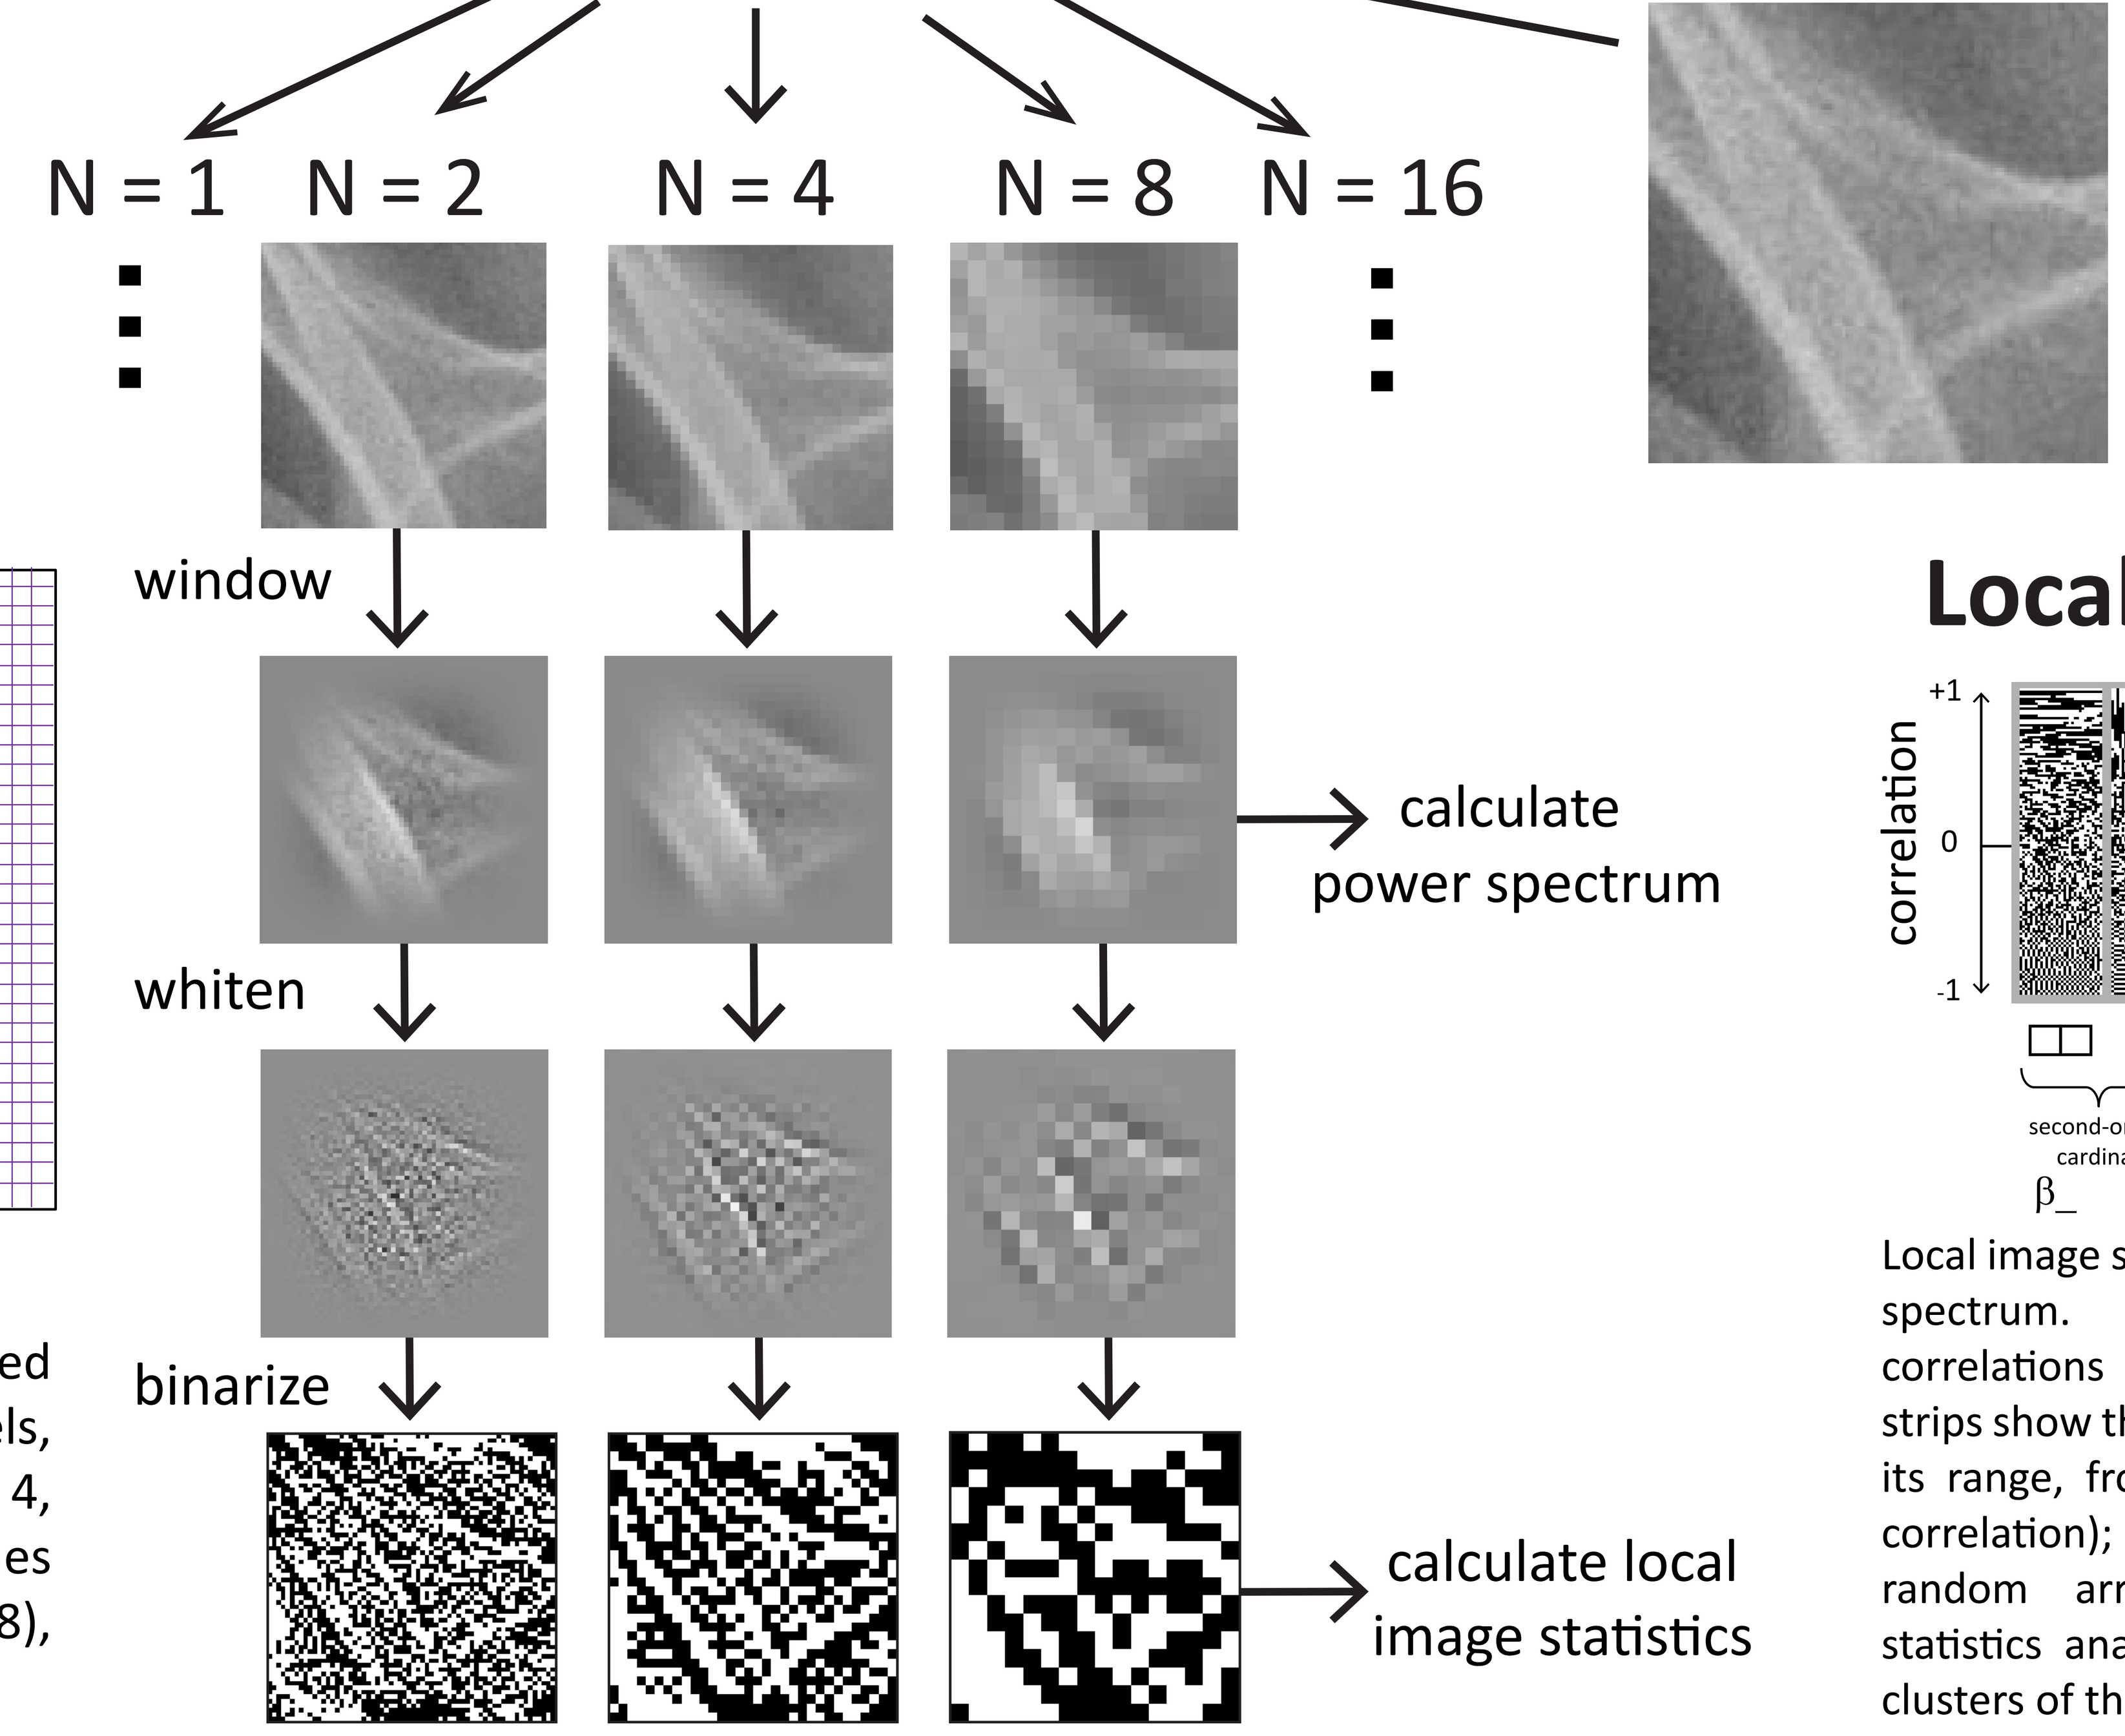

N = 1
N = 2
N = 4
N = 8
N = 16
window
whiten
binarize
calculate power spectrum
calculate local image statistics

Local Image Statistics

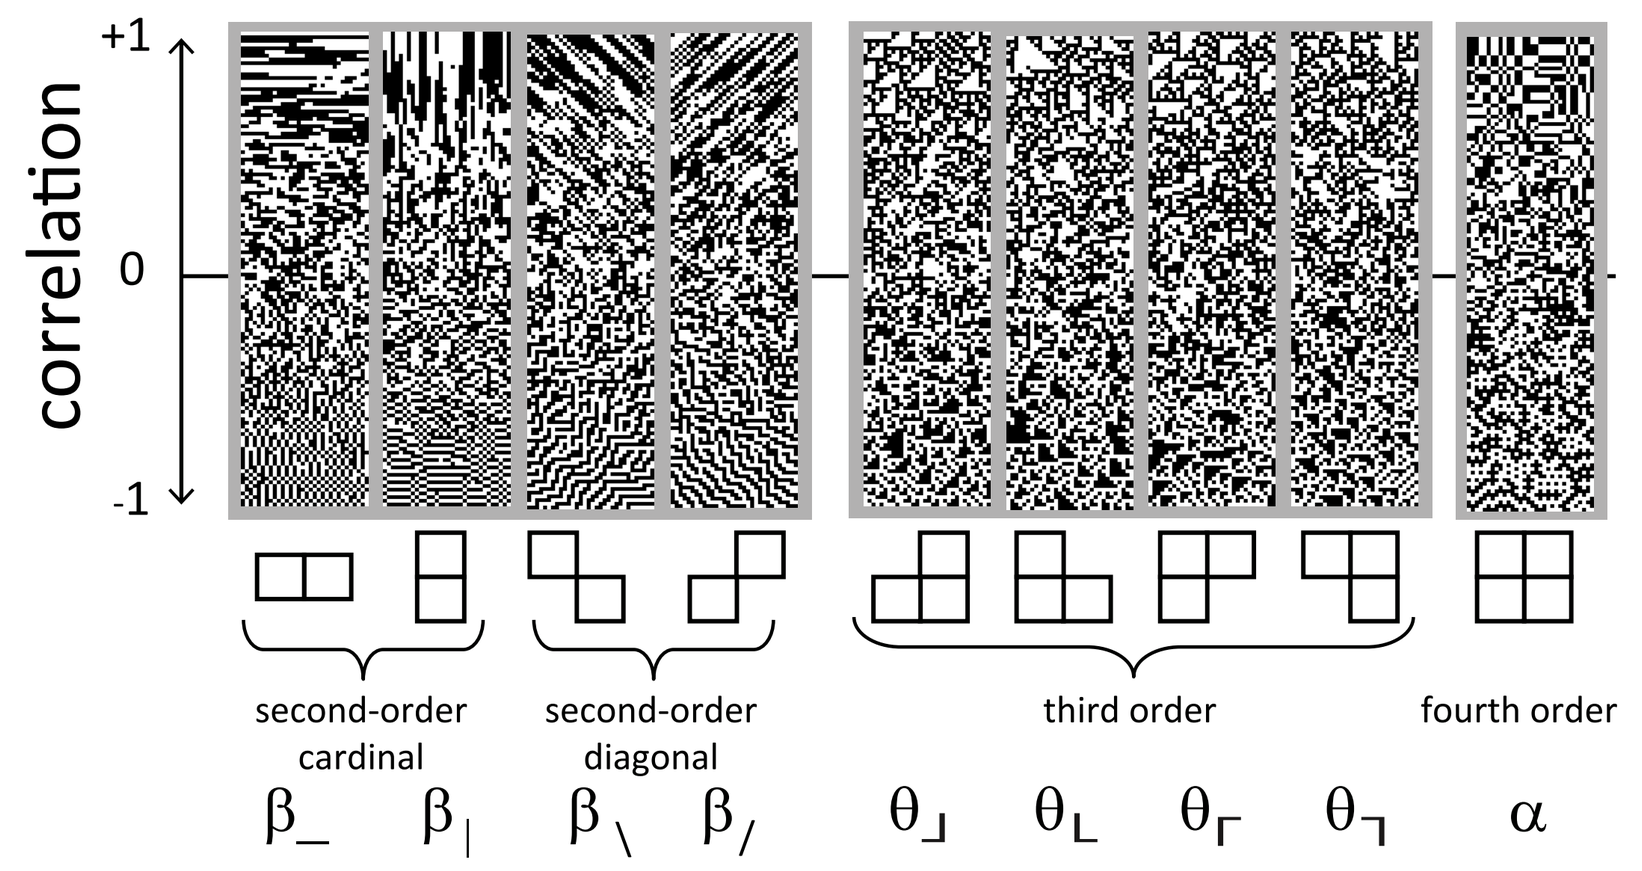

correlation
+1
0
-1
second-order cardinal
second-order diagonal
third order
fourth order

β_{-} $\beta_{|}$ $\beta_{\backslash}$ $\beta_{/}$ $\theta_{\lrcorner}$ $\theta_{\llcorner}$ $\theta_{\ulcorner}$ $\theta_{\urcorner}$ α

Local image statistics are complementary to the power spectrum. Second-order statistics describe correlations between pairs of adjacent checks. The strips show the effects of varying each statistic through its range, from -1 (anticorrelation) to +1 (complete correlation); 0 corresponds to no correlation (a random arrangement). Third- and fourth-order statistics analogously describe correlations between clusters of three and four checks.

Power Spectra

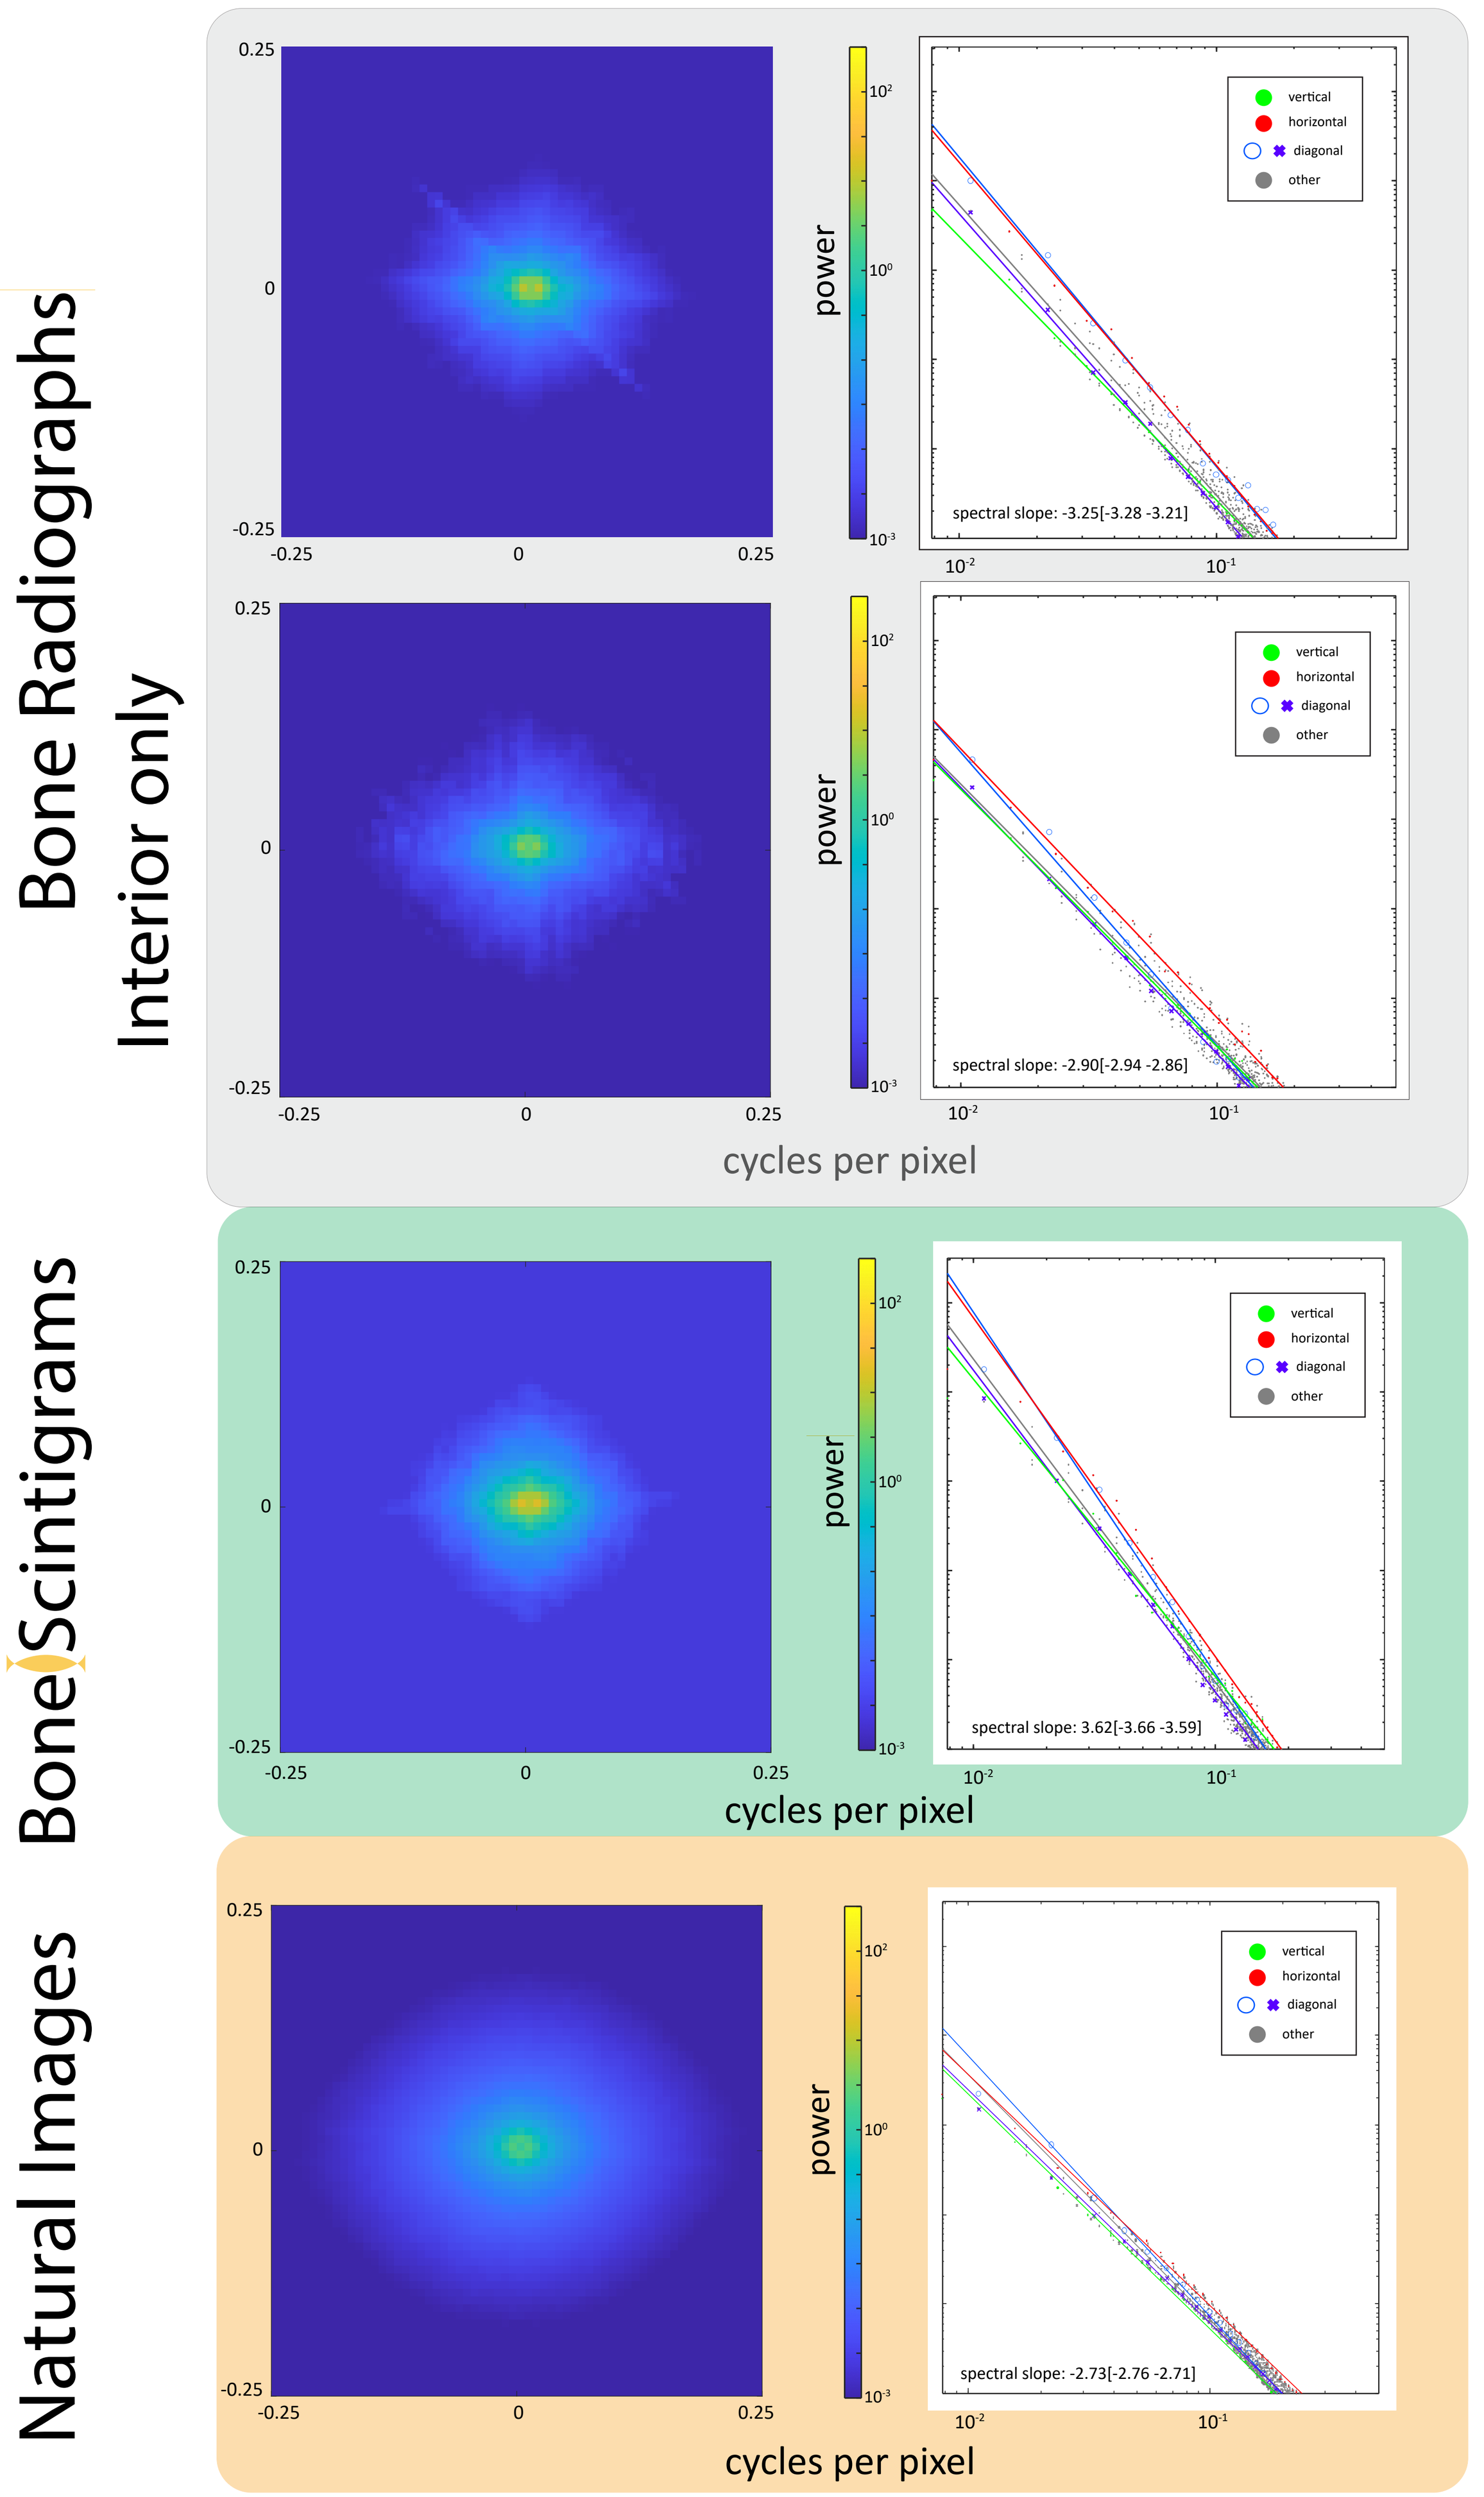

Bone Radiographs
Interior only
Bone Scintigrams
Natural Images
power
spectral slope: -3.25[-3.28 -3.21]
spectral slope: -2.90[-2.94 -2.86]
spectral slope: 3.62[-3.66 -3.59]
spectral slope: -2.73[-2.76 -2.71]
vertical
horizontal
diagonal
other
cycles per pixel

Results

Power Spectra

We characterized the statistics of each image set via their spectra and calculated the spectral slopes. Medical image sets have more anisotropy than natural images, as seen from the ridges in their power spectra. Some of the oriented ridges in the power spectra of radiographs are not present when only bone interiors are analyzed, indicating that they are caused by the sharp edges of bones.

Local Image Statistics

We computed the local image statistics of each image set and summarized them by their mean and standard deviation (across patches) at each scale (N: block size, R: patch size, in blocks). One difference is that for third-order statistics, the mean and standard deviations are smaller than for natural images. Error bars: 95% confidence limits.

Local Image Statistics

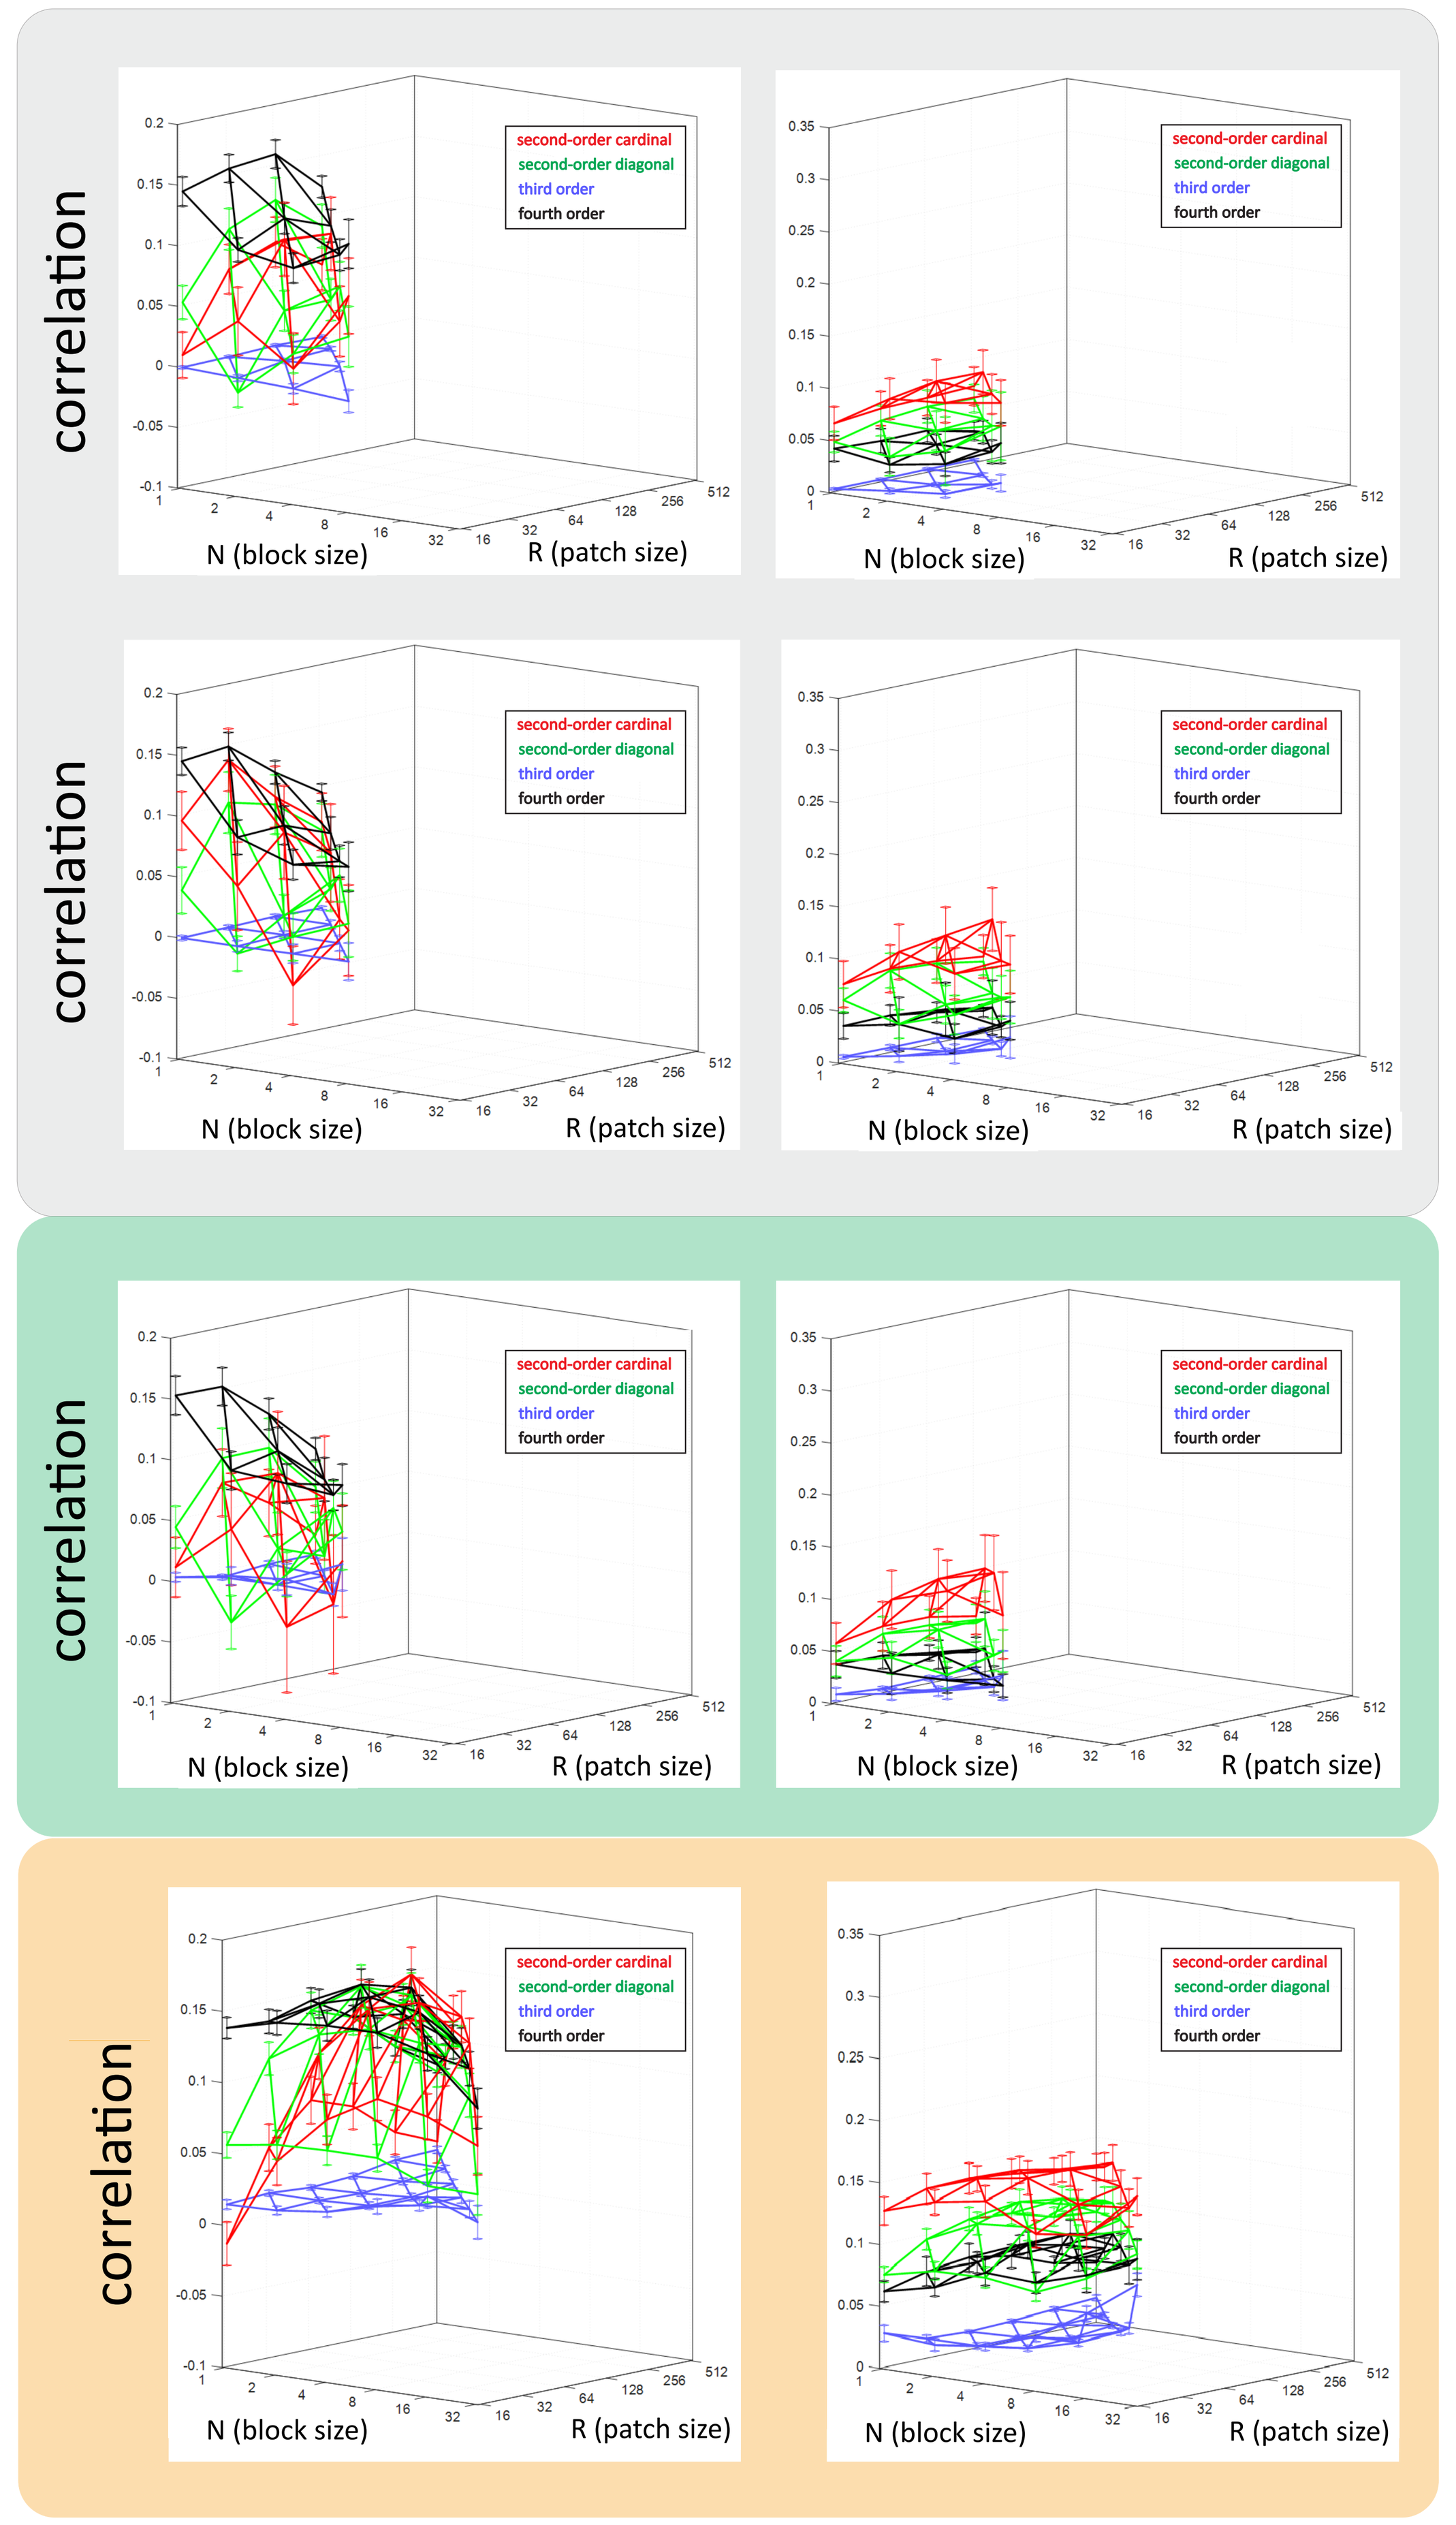

means
standard deviations
correlation
second-order cardinal
second-order diagonal
third order
fourth order
N (block size)
R (patch size)

Summary and Conclusions

Bone radiographs and scintigrams have statistical properties that differed from those of natural images.

- The spectral slopes of bone radiographs (-3.2 for raw images, -2.9 for interior) and of scintigrams (-3.6) were steeper than that of natural mages (-2.7, processed via the same pipeline).
- Local image statistics also differed in these medical images, compared to natural images.
 - Local correlations of all orders were less informative in medical images.
 - Third-order correlations were especially uninformative in medical images, a finding that we speculate may relate to the lack of T-junctions.

References

Field, D. J. (1987) Relations between the statistics of natural images and the response properties of cortical cells. J. Opt. Soc. Am. 4, 2379–2394.

Hermundstad, A.H., Briguglio, J.J., Conte, M.M, Victor, J.D., Balasubramanian, V., and Tkačik, G., (2014) Variance predicts salience in central sensory processing. eLife 2014;10.7554/eLife.03722.

Victor, J.D. and Conte, M.M. (2012) Local image statistics: maximum-entropy constructions and perceptual salience. J. Opt. Soc. Am. A, 29, 1313-1345.

Contact: Jonathan Victor at jdvicto@med.cornell.edu

Support: *CA241705, EY07977, and the Feil Family Brain and Mind Research Institute*